orladeyo®
(berotralstat) capsules 150 mg oral pellets 72·96·108·132 mg

biocryst®

About ORLADEYO® (berotralstat)

CONTENTS

- About ORLADEYO
- How ORLADEYO Works
- ORLADEYO Development and Availability
- Patient Impact and Access
- Clinical Data and Real-world Evidence
- Established Safety
- Important Safety Information

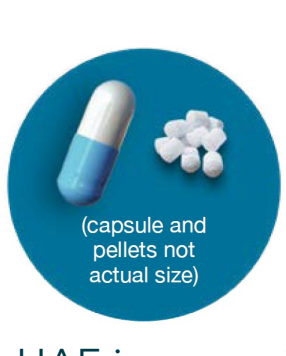

(capsule and pellets not actual size)

ORLADEYO® (berotralstat) is the first and only targeted preventative therapy for hereditary angioedema (HAE) to combine effective attack prevention and oral administration in adult and pediatric patients aged 2 years and older. ORLADEYO provides proven, significant and sustained attack prevention with once-daily oral administration.[1] It is available as a capsule for adults and pediatric patients 12 years and older and as oral pellets for pediatric patients aged 2 to <12 years.

HAE is a rare, inherited disease characterized by repeated painful and unpredictable episodes (or attacks) of severe submucosal and/or subcutaneous swelling in various parts of the body.[2] Approximately 40% of children experience their first HAE attack by age 5.

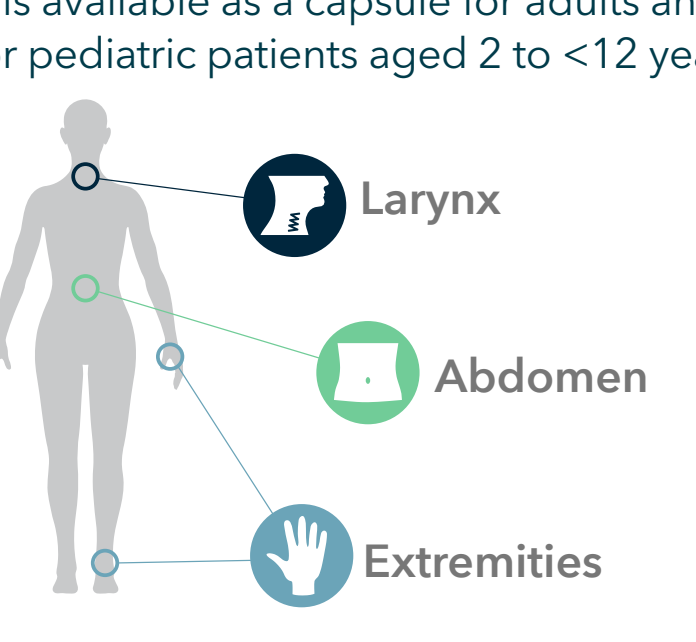

Larynx
Abdomen
Extremities

It affects about 1 in 50,000 people in the United States and, in most cases, is autosomal-dominant, meaning children of a parent diagnosed with HAE have a 50% chance of inheriting the disorder.[2]

HAE Treatment and the Impact of an Oral Option

HAE consensus guidelines state that the overall goals of HAE treatment are to reduce morbidity and mortality, and to restore a normal quality of life to patients.[3]

The treatment options available to people with HAE can be divided into two categories and should be individualized based on a patient's lifestyle, medical needs and preferences:[4]

- **On-demand therapy** is provided for acute HAE attacks and focuses on reducing the severity and duration of an ongoing attack, with the goal of resolving symptoms as quickly as possible.[4,5]
- **Prophylactic therapy** is designed to prevent attacks. The short-term goal of prophylactic therapy focuses on reducing the likelihood of swells in response to an anticipated traumatic event, like a medical procedure. Treatment with long-term prophylactic therapy focuses on reducing the number and severity of HAE attacks and associated burden of disease, and restoring normal quality of life. There are multiple routes of administration for preventative therapies, including intravenous, subcutaneous and oral.[3,4]
- **Some therapies can be burdensome.** For example, some intravenous therapies require injecting oneself up to 122 times per year for prophylaxis alone.[3] Treatments that are delivered intravenously or through subcutaneous injection (under the skin) can be inconvenient, burdensome and painful. A recent study showed that most people living with HAE would prefer an oral, once-daily preventative option, even if an injectable option meant less frequent dosing, as long as they could expect similar effectiveness.[a]

ORLADEYO offers:

Easy administration
One capsule or packet of oral pellets once per day with a meal

Convenient storage
Room temperature; no refrigeration needed

Flexible dosing
ORLADEYO can be taken anywhere

Innovative design
Not dependent on the availability of human plasma

[a]According to an online survey of 150 people aged ≥18 years living with HAE. Ninety-eight percent of respondents were receiving HAE treatment at the time of the survey. Dosing frequency did not significantly impact respondents' answers for preferring oral prophylaxis.

Back to CONTENTS >

SELECT IMPORTANT SAFETY INFORMATION
It is not known if ORLADEYO is safe and effective to treat an acute HAE attack, therefore ORLADEYO should not be used to treat an acute HAE attack. Do not take more than 1 dose of ORLADEYO per day because extra doses can cause heart rhythm problems.

Please see Important Safety Information at the bottom and full Prescribing Information, including the Patient Information, at Orladeyo.com.

How ORLADEYO Works

One dose of ORLADEYO per day works to prevent HAE attacks by decreasing the activity of plasma kallikrein.

Untreated HAE

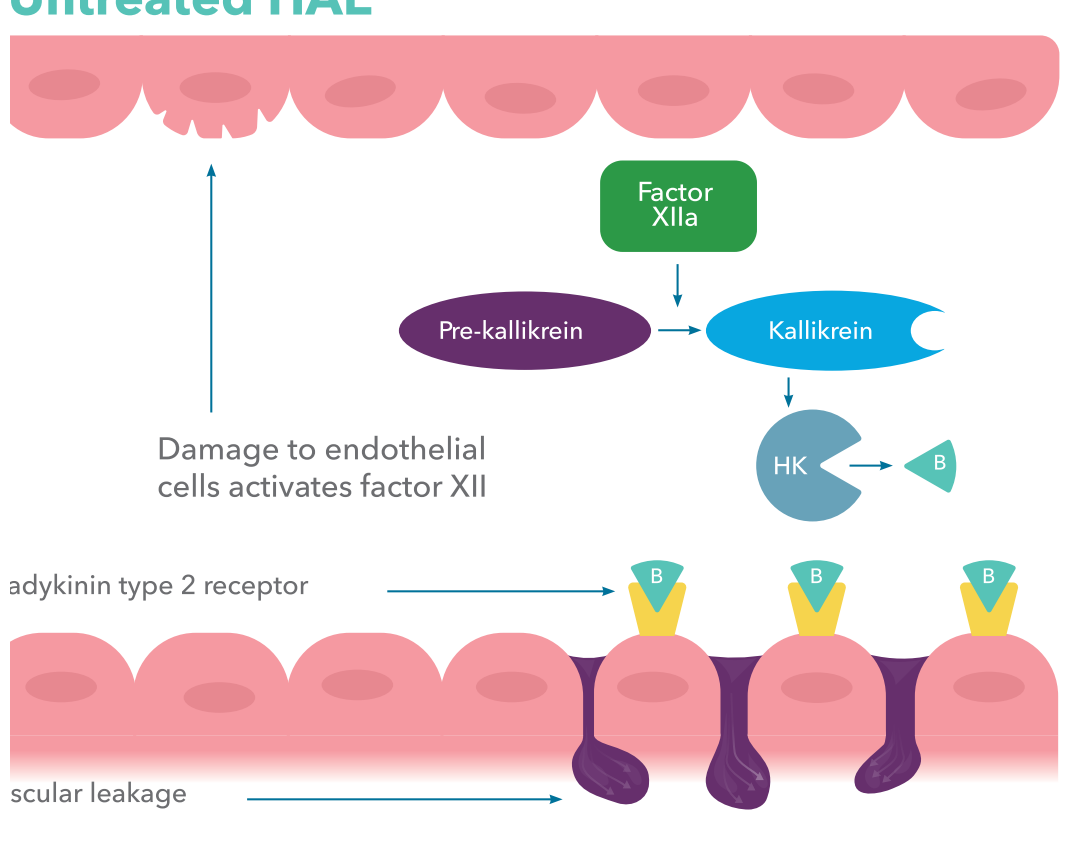

Factor XIIa
Pre-kallikrein
Kallikrein
HK
B
Damage to endothelial cells activates factor XII
adykinin type 2 receptor
scular leakage

With ORLADEYO

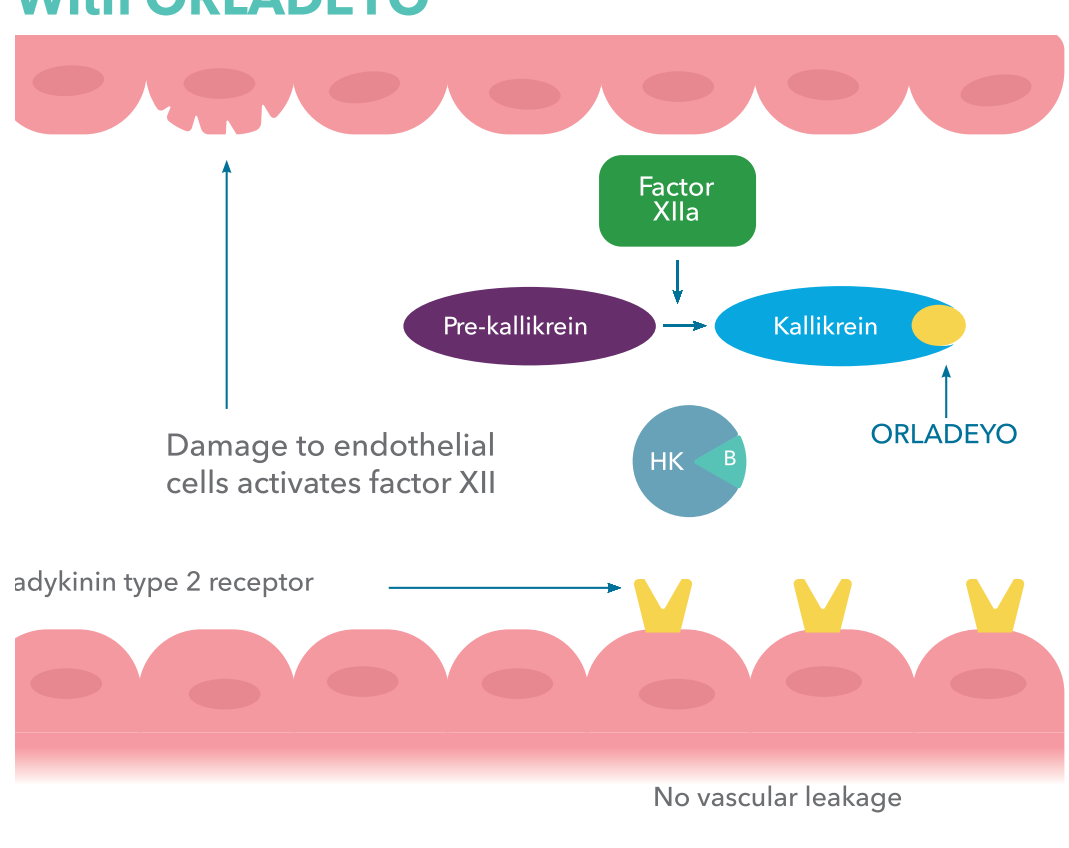

Factor XIIa
Pre-kallikrein
Kallikrein
ORLADEYO
HK
B
Damage to endothelial cells activates factor XII
adykinin type 2 receptor
No vascular leakage

Abbreviations: B, bradykinin; factor XIIa, activated factor XII; HK, high-molecular-weight kininogen.

In HAE, uncontrolled plasma kallikrein activity triggers an overproduction of bradykinin, which leads to vasodilation, vascular leakage and subsequent swelling.[6,7]

By decreasing plasma kallikrein activity, ORLADEYO prevents the cleavage of HK and subsequent release of bradykinin, ultimately preventing HAE attacks.[1,6,7]

Back to CONTENTS >

ORLADEYO Development and Availability

BioCryst discovered ORLADEYO in 2013 and brought it to patients in just seven years. In December 2020, a capsule formulation of ORLADEYO was approved by the U.S. Food and Drug Administration (FDA) for adults and pediatric patients aged 12 and older, with many subsequent ex-U.S. approvals thereafter. In December 2025, an oral pellet formulation of ORLADEYO was approved by the FDA for pediatric patients aged 2 to <12 years.

Major ORLADEYO Milestones:

2012
- BioCryst announced a focus on rare diseases, including HAE

2013
- Discovered ORLADEYO (BCX7353) at the BioCryst Discovery Center of Excellence in Hoover, Ala.

2014
- Initiated Phase 2 APeX-1 clinical study

2018
- Received Fast Track Designation from FDA
- *New England Journal of Medicine* published APeX-1 results
- Initiated Phase 3 APeX-2 clinical study

2019
- Announced APeX-2 results
- Submitted New Drug Application (NDA) for a capsule formulation of ORLADEYO for patients 12 and older

2020
- Received FDA approval for a capsule formulation for patients 12 and older and launched in the US

2022
- Initiated APeX-P study in patients 2 to <12 years

2025
- NDA accepted for the oral pellet formulation of ORLADEYO for patients 2 to <12 years
- Filed applications for use in Europe and Japan for ages 2 to <12; additional global filings planned
- *Annals of Asthma, Allergy & Immunology* published APeX-P results
- Sold European ORLADEYO business to Neopharmed Gentili
- Received FDA approval for the oral pellet formulation in patients 2 to <12 years

TODAY
- The capsule formulation is now approved in more than 45 countries around the world

Back to CONTENTS >

SELECT IMPORTANT SAFETY INFORMATION
Before taking ORLADEYO, tell your healthcare provider about all of your medical conditions, including if you:
- have liver problems or kidney problems.
- are pregnant or planning to become pregnant. It is not known if ORLADEYO can harm your unborn baby.
- are breastfeeding or plan to breastfeed. It is not known if ORLADEYO passes into your breastmilk. Talk to your healthcare provider about the best way to feed your baby while taking ORLADEYO.

Please see Important Safety Information at the bottom and full Prescribing Information, including the Patient Information, at Orladeyo.com.

Patient Impact and Access

>3.5K
PATIENTS PRESCRIBED ORLADEYO*
SINCE DECEMBER 2020

~200
ANTICIPATED NET NEW PATIENTS
EACH YEAR FOR THE NEXT FEW YEARS

EMPOWER PATIENT SERVICES

BioCryst's Empower Patient Services is a single-source solution for all patient and HCP ORLADEYO needs. It combines a specialty pharmacy and patient support program with a dedicated team of experts who provide individualized and streamlined support every step of the way.

*as of 3Q25

Back to CONTENTS >

Clinical Data and Real-world Evidence

The safety and effectiveness of ORLADEYO has been evaluated in a robust clinical program – including the APeX-S and APeX-2[a] studies in patients age 12 and older with confirmed HAE type 1 or 2 – and its real-world clinical use has been analyzed as part of BioCryst's continued commitment to demonstrating the impact of ORLADEYO.

Highlights from Final Published Analysis of APeX-2 96-week Data[9]

- Patients who received ORLADEYO 150 mg in this long-term, three-part study (n=26) reported rapid and sustained improvements from baseline of 3.3 attacks per month to 0.3 attacks per month after two years.
- Patients who received ORLADEYO 150 mg from Day 1 and completed the study (n=21) experienced an average reduction in monthly attack rate of 90.8% compared to baseline.
- More than half of people had zero attacks in 17 of the last 18 months of treatment. The primary objective in Part 3 of the study was to evaluate long-term safety and tolerablity.

Patients Experience Excellent HAE Control on ORLADEYO

LONG-TERM CLINICAL EVIDENCE[9]

90.8%
Attack reduction vs baseline after 96 weeks on ORLADEYO 150 mg in APeX-2

Source: Kiani-Alikhan S, Gower R, Craig T et al. Once-daily oral berotralstat for long-term prophylaxis of hereditary angioedema: The open-label extension of the APeX-2 randomized trial. J Allergy Clin Immunol December 2023

LONG-TERM REAL-WORLD EVIDENCE[10,11]

RAPID
attack control regardless of baseline rate or C1-INH level and function

SUSTAINED
attack control regardless of baseline rate or C1-INH level and function

ZERO
median monthly attack rate on ORLADEYO for patients who were attack free at baseline

Source: AAAAI Annual Meeting 2025 Poster Presentations, February 2025

[a]APeX-2 was a 3-part Phase 3 study. Participants included 121 patients aged 12 years and older with confirmed HAE type 1 or 2 who experienced ≥2 investigator-confirmed attacks during the 8-week run-in period. In APeX-2 Part 1, AE-QoL (secondary endpoint) scores meaningfully improved in all study arms beyond the AE-QoL MCID of 6; however, change in AE-QoL total scores from baseline to week 24 was not significant versus placebo.

Data Highlights

BioCryst has reported other long-term data and additional analyses from ORLADEYO clinical studies and real-world evidence, including:

CLINICAL RESULTS

- Results from Part 3 of APeX-2 published in *JACI: In Practice* showed clinically meaningful reductions in HAE attacks and improved quality of life[a] over 96 weeks of treatment with ORLADEYO.[9]
- Final safety and effectiveness analysis from APeX-S showed ORLADEYO 150 mg resulted in a median attack rate of 0.0 attacks per month in 20 of 24 months (AAAAI 2023).[12]
- Patients remained attack free for 98% of days after switching from an injectable therapy to ORLADEYO (*Annals of Allergy, Asthma & Immunology*, 2024).[13]
- Patients who completed 96 weeks on ORLADEYO 150 mg in APeX-2 saw an 88% reduction in rescue medication use,[b] and had 95% attack-free days in Part 3 of the study.[9]
- One-year data from APeX-P, the largest HAE long-term prophylactic trial in patients 2 to <12 years of age, showed that ORLADEYO demonstrated an early and sustained reduction in attacks, with a median attack rate of 0 for 11 out of 12 months, beginning at month 1 (ACAAI 2025).[14]

[a]Hypothesis testing and statistical analyses were not prespecified in the protocol for Part 3, and as such conclusions around the significance of changes from baseline cannot be made.
[b]At 2 years of treatment with ORLADEYO 150 mg compared with baseline (n=19 patients; 2 patients did not receive any rescue medication at baseline)

REAL-WORLD EVIDENCE

- Real-world evidence showed reductions in hospitalizations and emergency room/clinician visits among U.S. patients following initiation of ORLADEYO (*Journal of Managed Care and Specialty Pharmacy*, 2025).[15]
- Real-world data showed patients 12 and above who initiated ORLADEYO experienced substantial and sustained reductions in attack rates through 18 months of treatment regardless of HAE type, baseline attack frequency, and prior long-term prophylaxis (AAAAI 2025).[10,11]
- Real-world evidence showed high adherence and persistence rates among patients 12 and above treated with ORLADEYO that were equivalent to – and in some cases numerically higher than – other long-term prophylactic therapies (*Allergy and Asthma Proceedings*, 2025).[16]

Back to CONTENTS >

Established Safety

The safety of ORLADEYO in patients 12 years and older is supported by data from patients across two clinical studies in 508 patients and analyses of real-world outcomes.[1,12,13]

In APeX-2 Part 1, the most common[a] treatment-emergent adverse reactions (TEAEs) were abdominal pain, vomiting, diarrhea, back pain and gastroesophageal reflux disease (GERD).[1] Gastrointestinal (GI) adverse reactions generally occurred early after initiation of treatment, became less frequent with time and typically self-resolved.

	n (%)		
Adverse reactions	**Placebo** (n=39)	**ORLADEYO 110 mg** (n=41)	**ORLADEYO 150 mg** (n=40)
Abdominal pain[b]	4 (10)	4 (10)	9 (23)
Vomiting	1 (3)	4 (10)	6 (15)
Diarrhea[c]	0	4 (10)	6 (15)
Back pain	1 (3)	1 (2)	4 (10)
GERD	0	4 (10)	2 (5)

[a] ≥10% and higher than placebo.[1]
[b] Includes abdominal pain, abdominal discomfort, abdominal tenderness and upper abdominal pain.[1]
[c] Includes diarrhea and frequent bowel movements[1]

No patients in the ORLADEYO 150 mg dose group and one patient in the ORLADEYO 110 mg dose group discontinued treatment due to a GI adverse reaction in APeX-2 Part 1.[1]

Findings from the open-label, long-term safety study, APeX-S (final safety population, n=387), support the data observed in APeX-2 Part 1.[1,13,17]

No new safety signals were seen in those who continued ORLADEYO for 96 weeks. In APeX-2 Part 3 (n=81), the most common TEAEs were nasopharyngitis, urinary tract infection, abdominal pain, arthralgia, coronavirus infection and diarrhea. One patient receiving ORLADEYO 150 mg discontinued treatment due to a GI abdominal adverse reaction in APeX-2 Part 3.[9]

In APeX-S (n=387), the switch from lanadelumab to ORLADEYO was generally well tolerated. The most common adverse events (occurring in ≥5% of patients) were vomiting, diarrhea, upper respiratory tract infection, nausea, insomnia, pharyngitis, abdominal discomfort, back pain and dysmenorrhea. No patients in this analysis discontinued ORLADEYO due to an adverse event.[13]

Findings from APeX-P (n=29) demonstrate the safety and tolerability of ORLADEYO in patients 2 to <12 years of age, with the most common TEAEs being nasopharyngitis (34.5%), upper respiratory tract infection (27.6%), vomiting (17.2%), and headache (13.8%).[14]

Back to CONTENTS >

INDICATION AND IMPORTANT SAFETY INFORMATION

WHAT IS ORLADEYO® (berotralstat)?

ORLADEYO (or-luh-DAY-oh) is a prescription medicine used to prevent attacks of hereditary angioedema (HAE) in adults and children 2 years of age and older. It is not known if ORLADEYO is safe and effective in children under 2 years of age.

It is not known if ORLADEYO is safe and effective to treat an acute HAE attack, therefore ORLADEYO should not be used to treat an acute HAE attack.

Do not take more than 1 dose of ORLADEYO per day because extra doses can cause heart rhythm problems.

Before taking ORLADEYO, tell your healthcare provider about all of your medical conditions, including if you

- have liver problems or kidney problems.
- are pregnant or planning to become pregnant. It is not known if ORLADEYO can harm your unborn baby.
- are breastfeeding or plan to breastfeed. It is not known if ORLADEYO passes into your breastmilk. Talk to your healthcare provider about the best way to feed your baby while taking ORLADEYO.

Tell your healthcare provider about all of the medicines you take, including other medicines for HAE, prescription and over-the-counter medicines, vitamins, and herbal supplements.

Taking ORLADEYO with certain other medicines may affect the way other medicines work and other medicines may affect how ORLADEYO works.

Know the medicines you take and keep a list of them to show your healthcare provider and pharmacist when you get a new medicine.

What are the possible side effects of ORLADEYO?

Taking more than 1 dose of ORLADEYO per day may cause serious side effects, including **heart rhythm problems**. A heart rhythm problem called QTc interval prolongation can happen in people who take more than 1 dose of ORLADEYO per day. This condition can cause an abnormal heartbeat. Do not take more than 1 dose of ORLADEYO per day.

The most common side effects of ORLADEYO include abdominal pain, vomiting, diarrhea, back pain, and heartburn. These are not all of the possible side effects of ORLADEYO. For more information, ask your healthcare provider or pharmacist.

Talk to your healthcare provider for medical advice about side effects.

You are encouraged to report side effects of prescription drugs to BioCryst Pharmaceuticals, Inc. at 1-833-633-2279 or to the FDA at www.fda.gov/medwatch or 1-800-FDA-1088.

For more information, please see the full Prescribing Information including the Patient Information.

Back to CONTENTS >

You are encouraged to report side effects of prescription drugs to BioCryst Pharmaceuticals, Inc at 1-833-633-2279 or to the FDA at www.fda.gov/medwatch or 1-800-FDA-1088.

References **1.** ORLADEYO [prescribing information]. Durham, NC: BioCryst Pharmaceuticals Inc.; 2025. **2.** Bernstein JA. Am J Manag Care. 2018;24(14 suppl):S292-S298. **3.** Busse PJ, Christiansen SC, Riedl MA, et al. J Allergy Clin Immunol Pract. 2021;9(1):132-150.e3. **4.** Craig T, Busse P, Gower RG, et al. Ann Allergy Asthma Immunol. 2018;121(6):673-679. **5.** Cicardi M, Bork K, Caballero T, et al. Allergy. 2012;67(2):147-157. **6.**Busse et al. N Engl J Med. 2020;382(12):1136-1148. **7.** Kaplan et al. Ann Allergy Asthma Immunol. 2010;104(3):193-204. **8.** Data on File, BioCryst Pharmaceuticals Inc. **9.** Kiani-Alikhan et al. J Allergy Clin Immunol Pract. 2024;12(3):733-743.e10. **10.** Tachdjian et al. Poster presented at: 2025 AAAAI Annual Meeting; February 28-March 3, 2025. **11.** Davis-Lorton et al. Poster presented at: 2025 AAAAI Annual Meeting; February 28-March 3, 2025. **12.** Aygören-Pürsün et al. Poster presented at: 2023 AAAAI Annual Meeting; February 24-27, 2023. **13.** Riedl et al. Ann Allergy Asthma Immunol. 2024;132(4):505-511.e1. **14.** Bernatoniene et al. Poster presented at: 2025 ACAAI Annual Meeting; November 6-10, 2025. **15.** Christiansen SC, Lopez-Gonzales L, MacKnight SD, et al. J Manag Care Spec Pharm. 2025;31(6):578-589. **16.** Zuraw B, Lopez-Gonzales L, Manjelievskaia J, et al. Allergy Asthma Proc. 2025;46(3):209-217. **17.** Aygören-Pürsün et al. Poster presented at: 2023 World Allergy Congress; December 1-3, 2023.

orladeyo®
(berotralstat) capsules 150 mg oral pellets 72·96·108·132 mg

BioCryst and ORLADEYO are registered trademarks of BioCryst Pharmaceuticals, Inc. © 2026 BioCryst Pharmaceuticals, Inc. All rights reserved. US.ORL.03004 2/26

biocryst®